Meal Pattern Analysis in Sprague-Dawley and Diet-Induced Obese Levin Rats Following Peripheral Administration of Exenatide

Julie K. Wilson, David G. Parkes, Christine M. Mack
Amylin Pharmaceuticals, Inc., San Diego, CA

Abstract

Exenatide is an incretin mimetic approved for adjunctive therapy in patients with type 2 diabetes. In addition to improving glycemic control, exenatide also produces long-term weight reduction in humans and preclinical models. As decreased food intake has been found to contribute to the observed reduction in body weight in preclinical models, the current experiment examined dark-cycle feeding patterns in two different strains of rats treated with exenatide. Male Sprague-Dawley (SPD) rats (mean body weight = 458 g) and Levin (inbred) diet-induced obese (DIO) rats (mean body weight = 554 g), both consuming a diet consisting of 32% fat kcal, received a single intraperitoneal injection of exenatide (5 μg/kg) or vehicle, for 3 consecutive nights, at the onset of the dark cycle. Meal size, latency to eat, meal duration, inter-meal interval (IMI) and satiety ratio (IMI/meal size) were measured for the first meal each night. The total number of dark-cycle meals was also assessed each night. Feeding pattern analysis (with Treatment, Strain and Night as independent variables) revealed a main effect of Treatment for meal size, demonstrating a decrease in food intake in exenatide versus control treated rats ($P < 0.05$). A Strain X Treatment interaction ($P < 0.05$) for meal size revealed a greater magnitude of effect of exenatide in Levin DIO compared to SPD rats. There was also a main effect of Treatment, and Strain X Treatment interaction, for latency to eat ($P < 0.05$). Latency to eat specifically increased with exenatide treatment in SPD rats ($P < 0.05$), but not Levin DIO rats. There were no significant effects observed for meal duration or the IMI. A main effect of Treatment for the satiety ratio revealed a greater level of satiety in exenatide treated rats, regardless of strain ($P < 0.05$). Lastly, there was no effect of Treatment on meal number; however, a Strain X Treatment interaction ($P < 0.05$) revealed decreased meal number with exenatide treatment in SPD rats ($P < 0.05$), and a trend towards an increase in Levin DIO rats ($P = 0.05$). In summary, decreased meal size and an increased satiety ratio during treatment with exenatide was consistently observed in both strains of rats. These data suggest that exenatide regulates food intake through mechanisms involved in satiety.

The Obesity Society (NAASO)
October 20-24, 2006, Boston, MA
Poster number: 648-P

Introduction

Exenatide is an incretin mimetic approved for adjunctive therapy in patients with type 2 diabetes. In addition to improving glycemic control, exenatide also leads to long-term weight reduction in humans.[1] As decreased food intake has been observed in both animal and human studies,[2,3] and found to contribute to the observed reduction in body weight in preclinical models,[3] the present study examined dark-cycle feeding patterns in two strains of rats treated with exenatide.

Methods

- Subjects were male Sprague-Dawley® rats (SPD) (mean body weight = 458 g) and Levin (inbred) diet-induced obese (DIO) (mean body weight = 554 g) rats fed pelleted chow (32% kcal from fat)
- Rats were habituated to the testing cages for 7 days, followed by 3 nights of vehicle injections (10 total days); rats were housed in the testing chambers throughout the habituation and test period
- On test day, SPD rats and DIO rats were administered a single intraperitoneal injection (IP) of exenatide (5 μg/kg) or vehicle (10% DMSO) at the onset of the dark cycle; this was repeated for 3 consecutive nights
- Food intake was measured at 5-second intervals by an automated food intake measuring system (BioDAQ®, Research Diets); body weight was recorded nightly
- Latency to feed, meal size, meal duration, inter-meal interval, and satiety ratio (inter-meal interval/meal size) were analyzed for the first meal on each night (minimum meal size = 0.2 g, inter-meal interval ≥15 min); the total number of dark-cycle meals was also assessed each night
- Data are presented as mean ± SEM; group differences were analyzed using one-way analysis of variance with Treatment, Strain, and Night as independent variables

Figure 1. Exenatide decreased dark-cycle feeding in both SPD and DIO rats

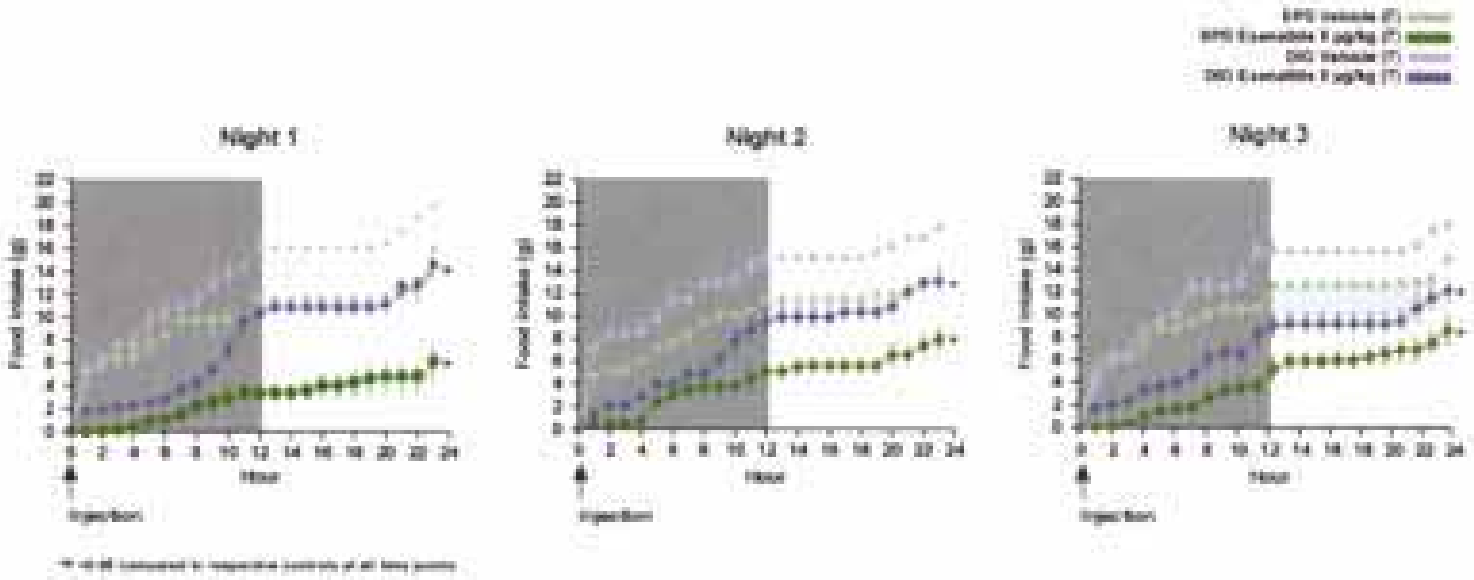
Night 1
Night 2
Night 3
Food Intake (g)
Hour
Injection

Percent Decrease in Food Intake at 12 h	SPD	DIO
Night 1	70%	36%
Night 2	55%	38%
Night 3	59%	43%

Figure 2. Exenatide produced a durable effect to decrease meal size in both SPD and DIO rats

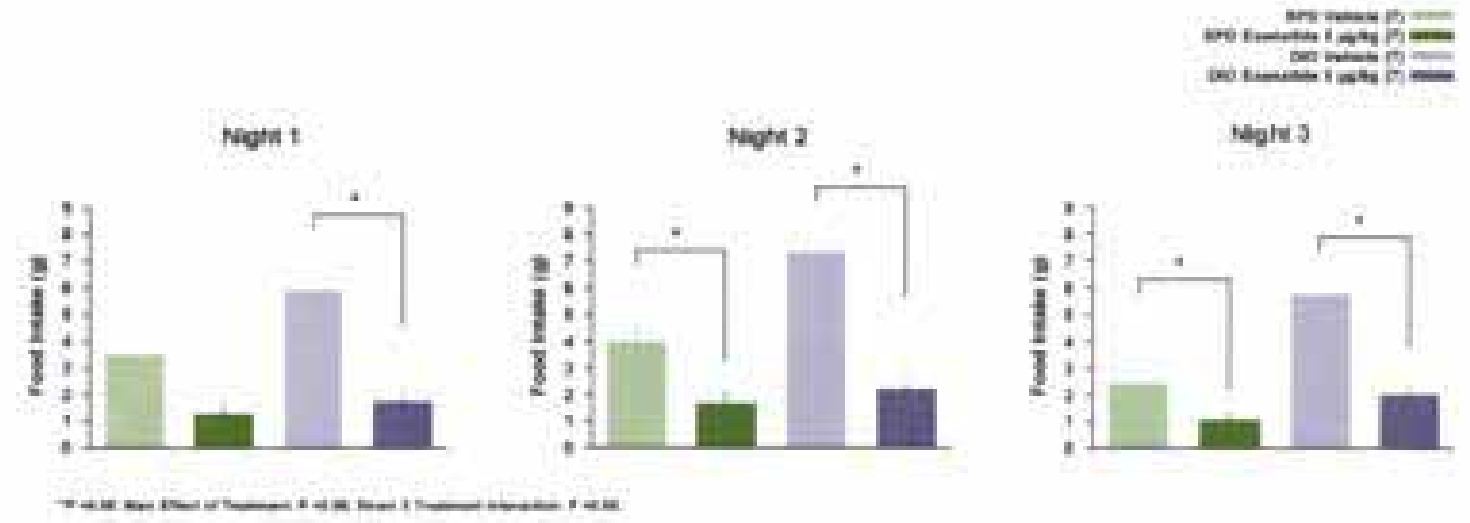
Night 1
Night 2
Night 3
Food Intake (g)

Figure 3. Exenatide reduced latency to feed in SPD but not DIO rats

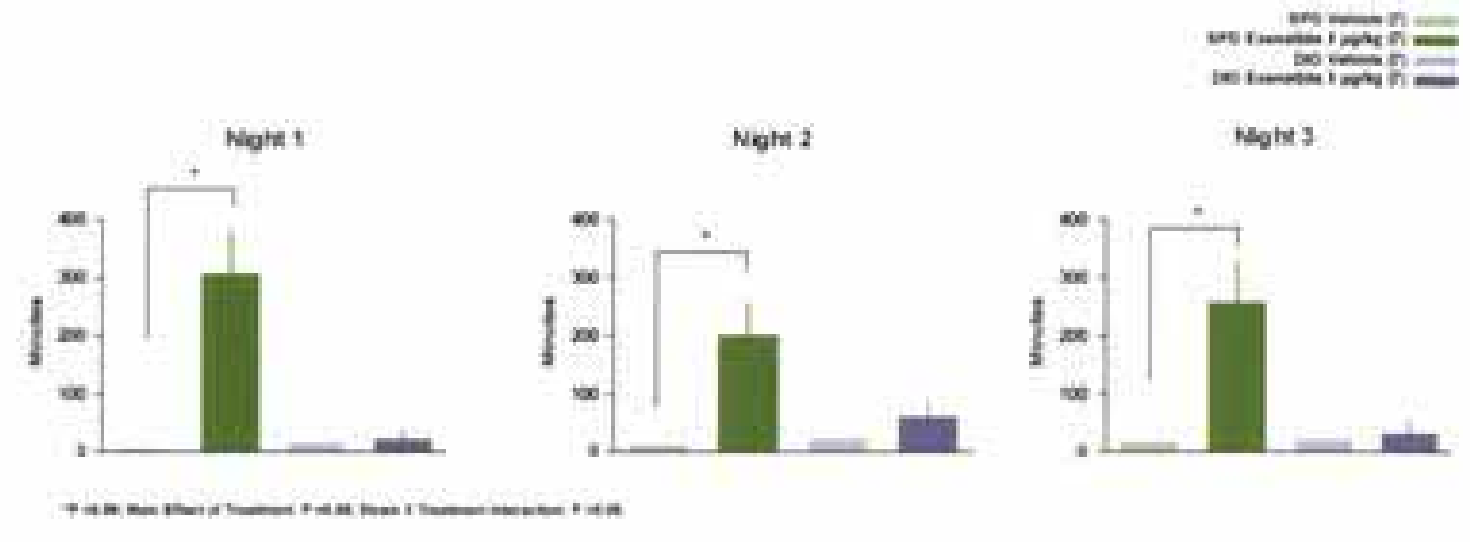
Night 1
Night 2
Night 3
Minutes

Figure 4. Exenatide had no effect on meal duration in SPD or DIO rats

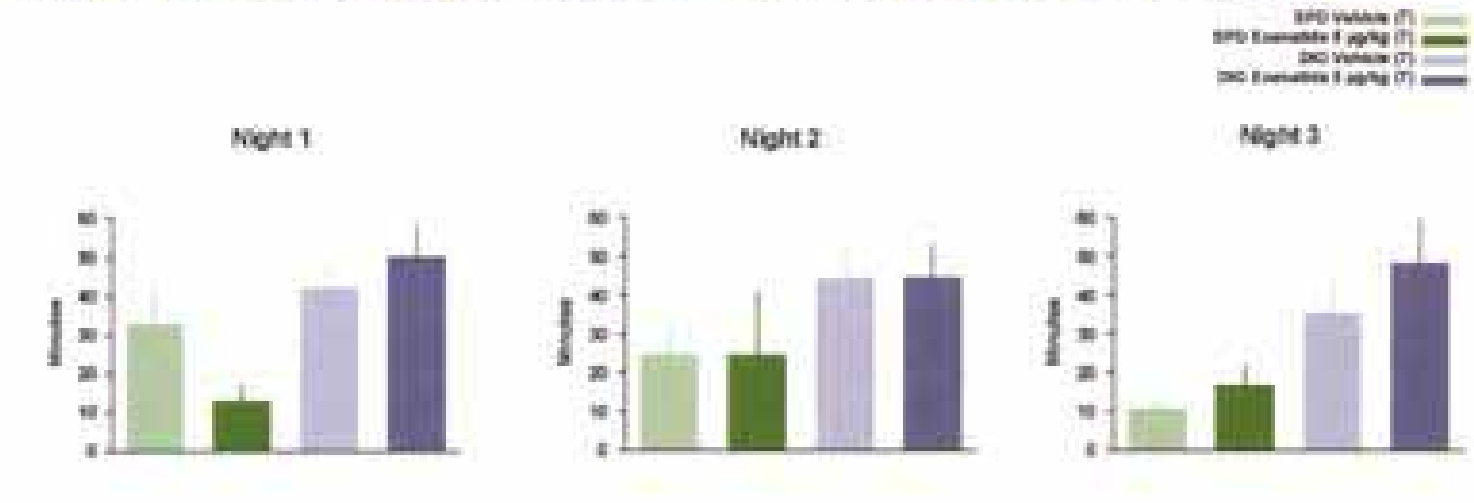
Night 1
Night 2
Night 3
Minutes

Figure 5. Exenatide had no effect on inter-meal interval in SPD or DIO rats

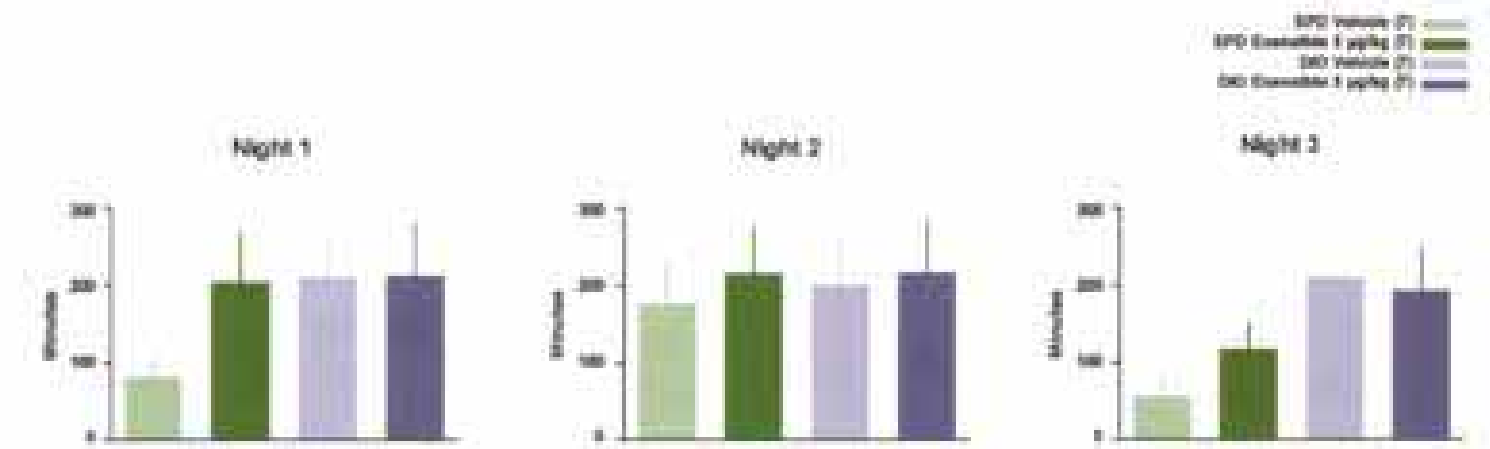
Night 1
Night 2
Night 3
Minutes

Figure 6. Exenatide increased the satiety ratio (inter-meal interval/meal size) in both SPD and DIO rats

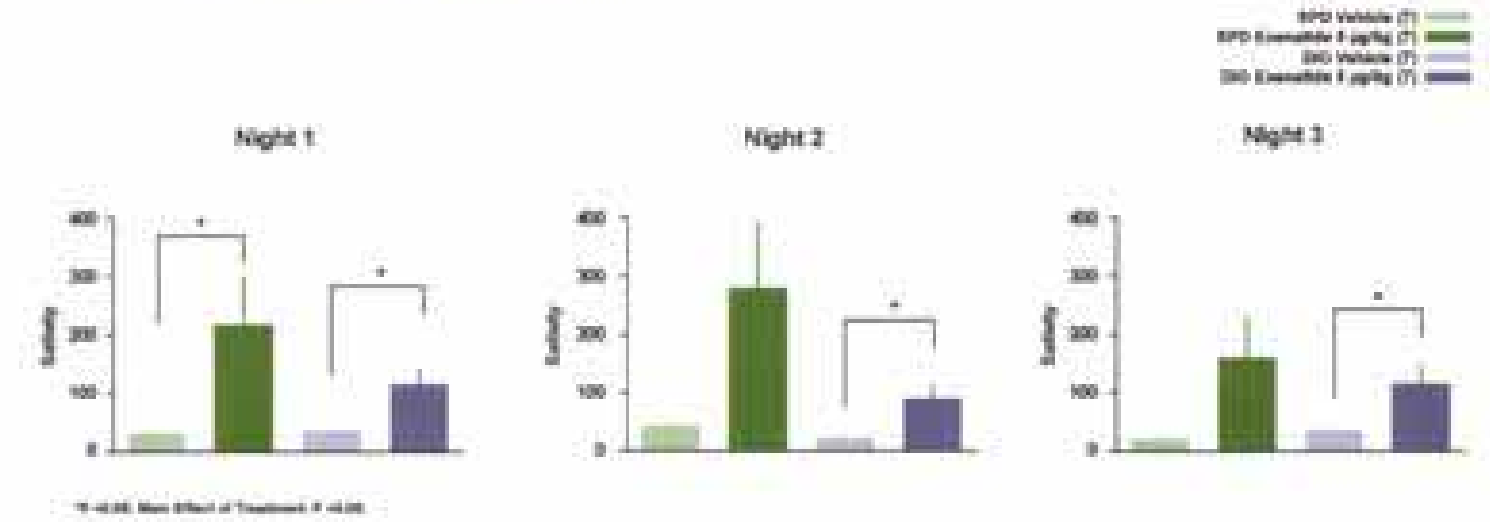
Night 1
Night 2
Night 3
Satiety

Figure 7. Number of dark-cycle meals was decreased in SPD rats, and increased in DIO rats, following exenatide treatment

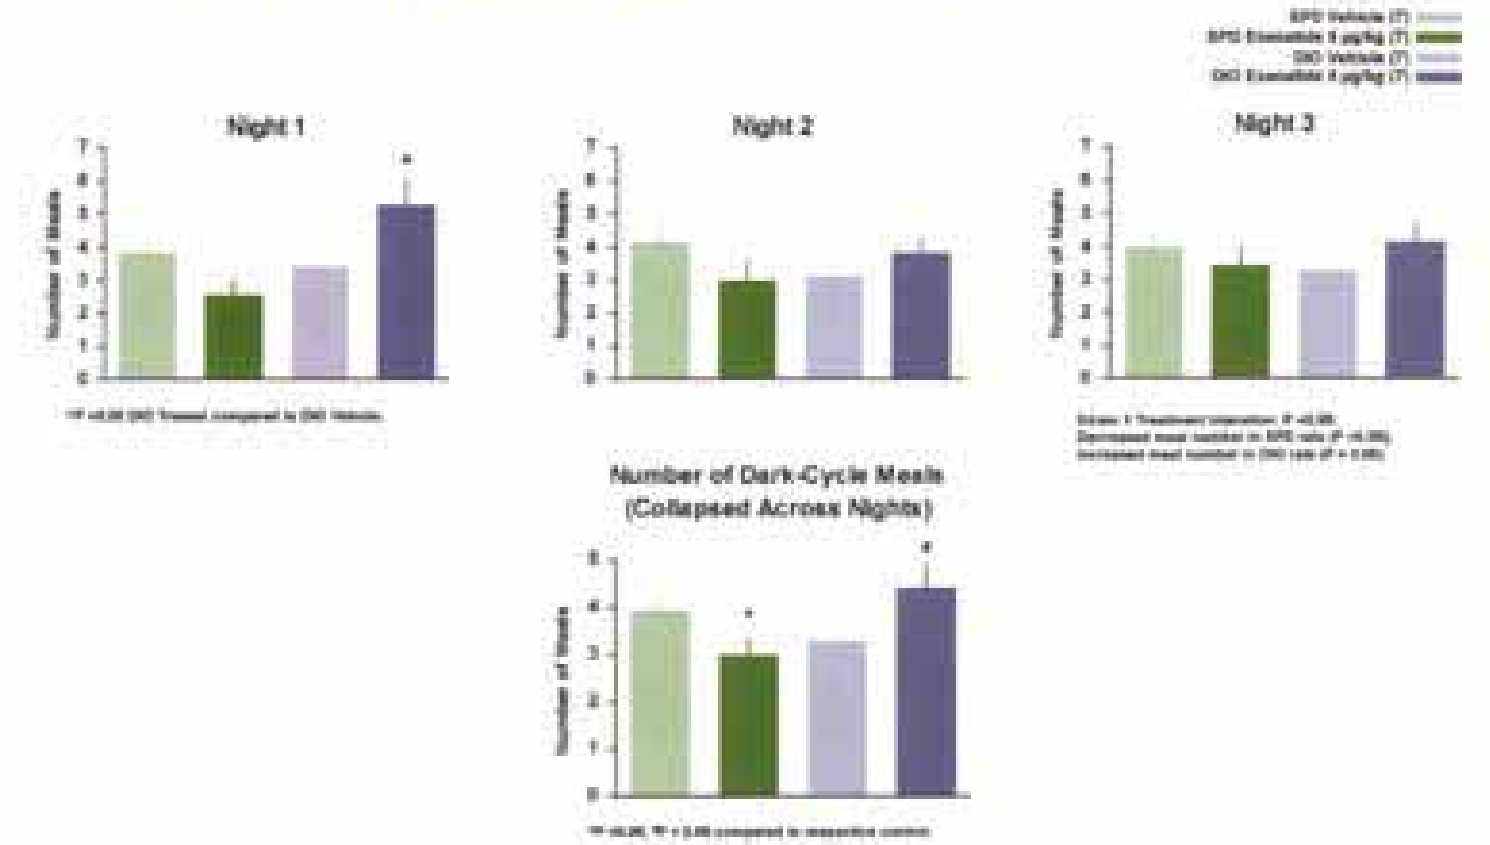
Night 1
Night 2
Night 3
Number of Meals
Number of Dark-Cycle Meals
(Collapsed Across Nights)

Figure 8. Exenatide decreased body weight in both SPD and DIO rats

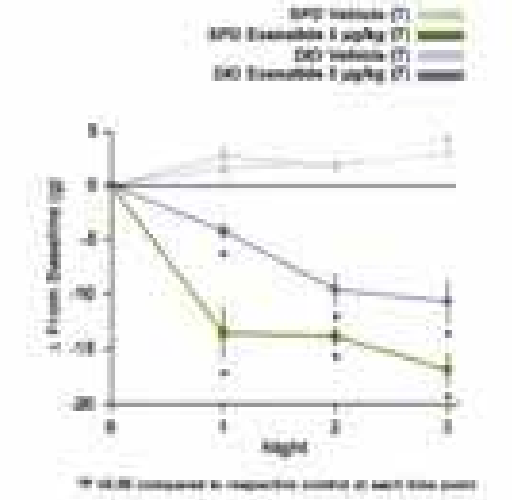
Night

Summary

- In both SPD and DIO rats, exenatide suppressed food intake compared to controls on each of the 3 nights 23 h post injection; this led to body weight loss in both strains at the end of treatment
- Exenatide increased latency to feed in SPD rats, but not DIO rats; the number of dark-cycle meals was decreased in SPD rats, and increased in DIO rats following exenatide treatment
- Exenatide had no effect on meal duration or inter-meal interval in either strain
- Consistent in both strains was reduced meal size and increased satiation with exenatide treatment
- The reliability of exenatide to reduce meal size and increase satiation across rat strains provide support for a role of exenatide in regulating food intake, and subsequent body weight, through mechanisms involved in satiety

References

1. Blonde L, Han J, Mac S, Poon T, Taylor K, Kim D. Exenatide (Exendin-4) Reduced A1C and Weight over 82 weeks in overweight patients with type 2 diabetes. Diabetes 2005; 54 (Suppl 1): A118. Abstract 477-P.
2. Edwards CM, Stanley SA, Davis R, Brynes AE, Frost GS, Seal LJ, Ghatei, MA, Bloom, SR. Exendin-4 reduces fasting and postprandial glucose and decreases energy intake in healthy volunteers. Am J Physiol Endocrinol Metab 2001; 281:E155-E161.
3. Mack CM, Moore CX, Jodka CM, Bhavsar S, Wilson JK, Hoyt J, Roan JL, Vu C, Laugero KD, Parkes DG, Young AA. Antiobesity action of peripheral exenatide (exendin-4) in rodents: effects on food intake, body weight, metabolic status and side-effect measures. Int J Obesity 2006; 30:1332-1340.